The Hidden Healthcare Resource Utilization (HCRU) Burden of Melanoma Recurrence in the US: A Nationally Representative Matched Cohort Analysis

Salome S. Samant, MBBS, MPH, DHSc[1]; Yong Zhu, PhD[2]; Nicole Engel-Nitz, PhD[2]; Sameer R. Ghate, PhD[1]; Dmitri O. Grebennik, MD[1]; Shujing Zhang, PhD[1]; Laura Becker, MS[2]; Arunima Sachdev, MA[2]; Shiqiang Li, MS[2]; Ahmad A. Tarhini, MD, PhD[3]

[1]Merck & Co., Inc., Rahway, NJ, USA; [2]Optum, Eden Prairie, MN, USA; [3]H. Lee Moffitt Cancer Center and Research Institute, Tampa, FL, USA

Background

- Resected stage IIB/C and stage III melanoma carries a high risk of recurrence, impacting clinical prognosis and healthcare resource utilization (HCRU) associated with the disease
- There is a need for updated evidence on the incremental HCRU associated with locoregional (LR) and distant (DR) recurrences versus no recurrence (NR) so as to fully characterize the value of preventing melanoma recurrence

Methods

- We conducted a retrospective matched-cohort analysis using a large US claims database linked to electronic health records (Optum Market Clarity database)
- Adults diagnosed with stage IIB–IIID cutaneous melanoma (July 1, 2019–May 31, 2025) and resected within 4 months were observed until death, loss of follow-up, or end of study period
- Patients with pregnancy, clinical trial participation, or other primary cancers were excluded
- Recurrence events were identified using a claims based on literature[1-4] and categorized as LR or DR
- Controls with NR were matched 1:1 using propensity scores (age group, gender, race/ethnicity, year of initial melanoma diagnosis, stage, baseline National Cancer Institute (NCI) Charlson comorbidity index etc.). NR_1 and NR_2 were the no-recurrence controls matched to DR and LR cohorts respectively and were assigned an index date to allow equal time from resection to recurrence as their match
- LR patients progressing to DR were included in both analyses but censored 30 days pre-DR in the LR HCRU analysis
- The HCRU was categorized by setting as ambulatory visits, emergency room (ER) visits, inpatient stays, and pharmacy fills. Pharmacy fills were based on pharmacy claims only; therapies administered under the medical benefit were captured under medical utilization
- All-cause and melanoma-related HCRU were reported per patient per month (PPPM) across all payers (**Figure 1**)

Figure 1. Patient selection and derivation of matched locoregional and distant recurrence cohorts among patients with resected stage IIB–IIID melanoma

N = 416,860
Evidence of cutaneous melanoma, defined as at least two non-diagnostic claims for melanoma in any position on different dates, during the identification period (July 1, 2019 to May 31, 2025). The earliest date of melanoma diagnosis is diagnosis date

↓

N = 157,710
Evidence of melanoma-related resection surgery within 120 days after diagnosis *and* age ≥ 18 years at surgery.The last surgery date within the120-day period is the index date (ID_1)

↓

N = 64,605
Continuous enrollment from 6 months before diagnosis to at least 1 month after index date

↓

N = 5,266
Patients with stage IIB-IIID, or proxy for stage 3[a]

↓

N = 5,105
No evidence of non-lymphnode metastasis prior to 90 days after index date

↓

N = 4,671
No hospice care or death within 90 days after index date and no pregnancy or clinical trial participation during the study period

↓

N = 3,218
No evidence of other primary cancer during the study period[b]

N = 1,000 patients with recurrence | **N = 2,218** patients without recurrence

1:1 Propensity score matching

608 pairs of patients with locoregional recurrence and matched controls
491 pairs of patients with distant recurrence and matched controls

[a]For Stage III Proxy: Have a lymph node (LN) excision surgery within 120 days following diagnosis and had no non-lymph node metastasis in the 2-months prior to diagnosis through 2-months following ID1 (resection) AND had LN metastasis in that period; [b]Except squamous cell or basal cell carcinomas or carcinomas in situ, for the entire study period from the beginning of baseline period to end of follow-up period or death.

Results

- After 1:1 propensity score matching, year 1 analyses included 608 LR–NR pairs and 491 DR–NR pairs; year 2 analyses included 155 LR–NR pairs and 112 DR–NR pairs
- Matching achieved strong balance in baseline characteristics for both LR vs NR and DR vs NR comparisons, with all matched covariates showing standardized mean difference <10%
- The mean age was 59.8 years in the LR cohort and 62.9 years in the DR cohort; 60.0% and 63.8% of patients were male, respectively, and most were non-Hispanic White
- Overall, most patients were classified as stage III using the claims-based proxy, including 93.3% of the LR cohort and 91.7% of the DR cohort. Additionally, stage IIB/C accounted for 2.1% of the LR cohort and 2.4% of the DR cohort, while documented stage III accounted for 4.6% and 5.9%, respectively

Figure 2. Incremental all-cause HCRU PPPM for LR and DR vs respective NR in year 1 and 2 after recurrence

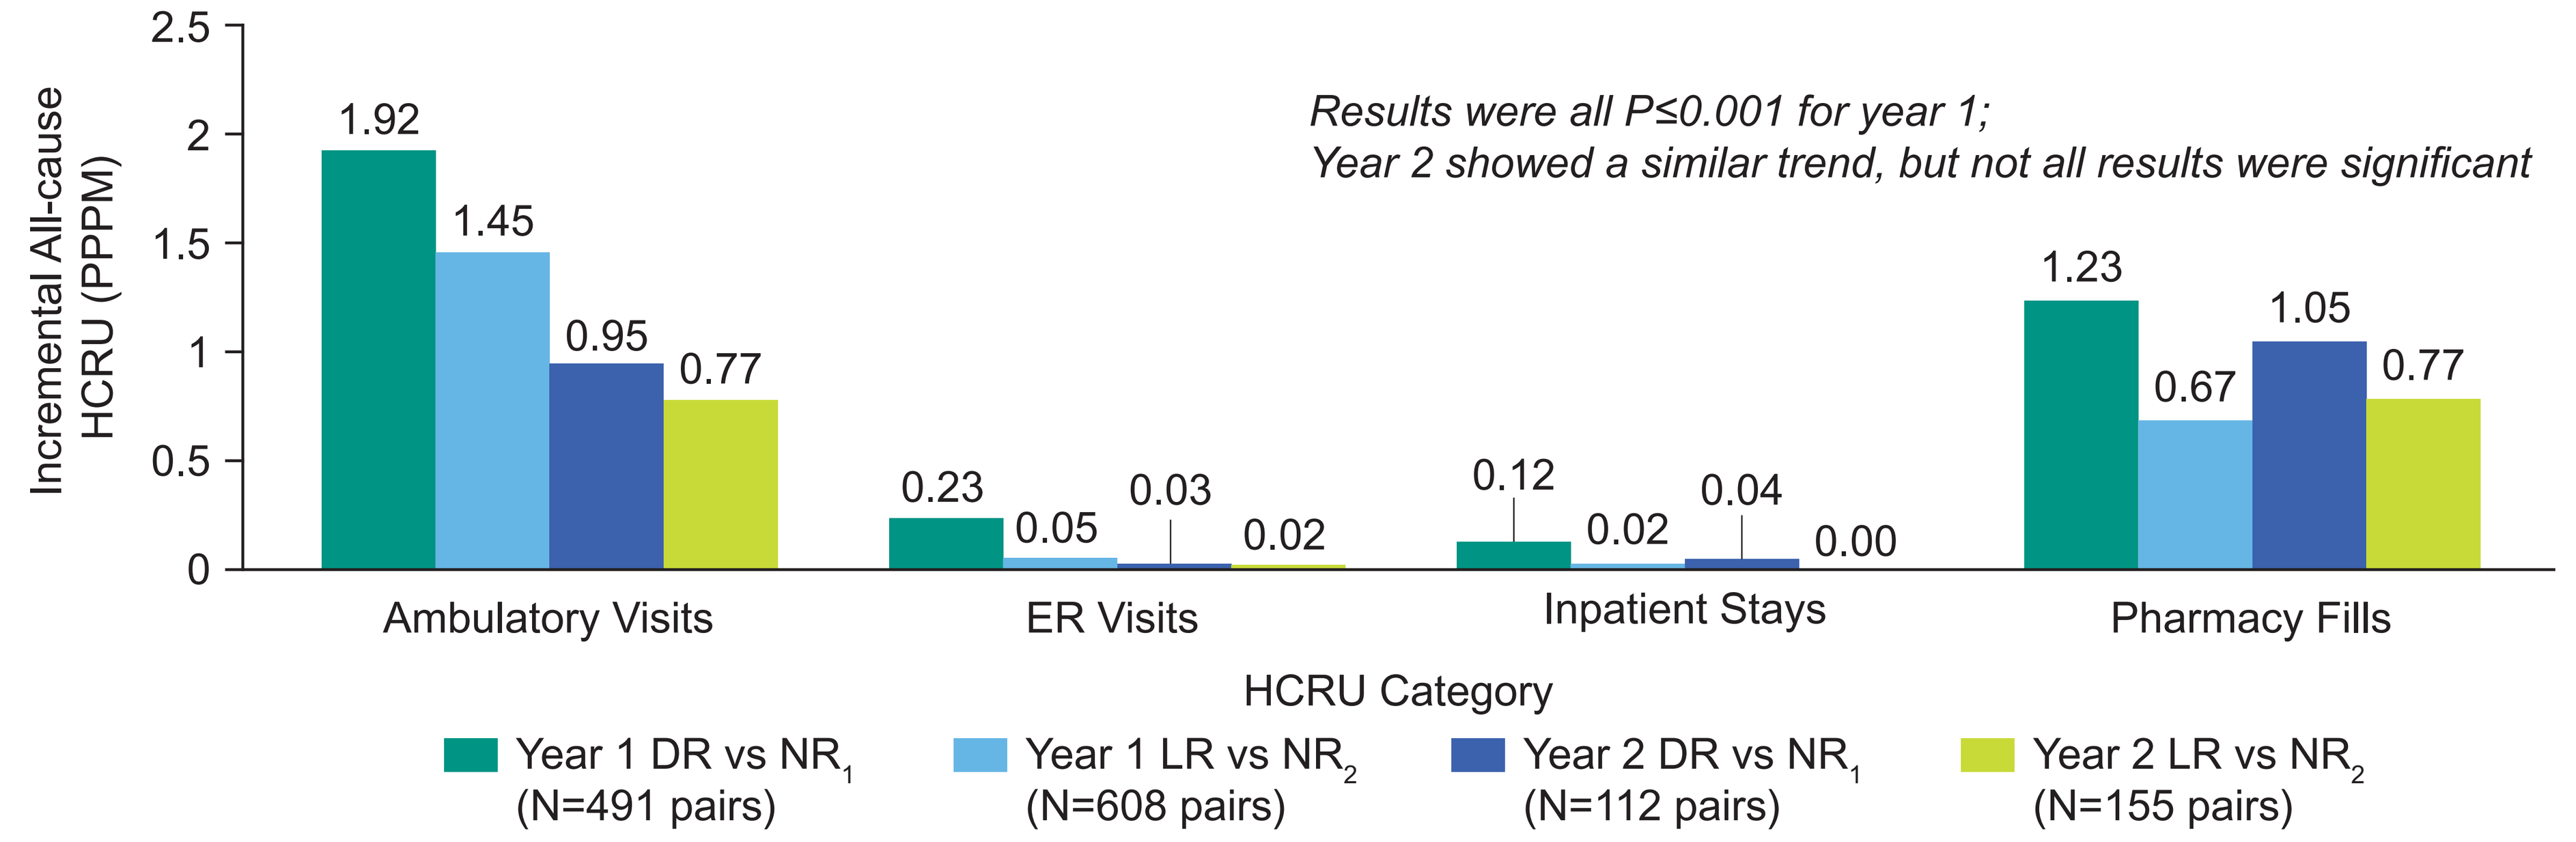
Incremental All-cause HCRU (PPPM)
2.5
2
1.5
1
0.5
0
1.92
1.45
0.95
0.77
0.23
0.05
0.03
0.02
0.12
0.02
0.04
0.00
1.23
0.67
1.05
0.77
Results were all P≤0.001 for year 1;
Year 2 showed a similar trend, but not all results were significant
Ambulatory Visits
ER Visits
Inpatient Stays
Pharmacy Fills
HCRU Category
Year 1 DR vs NR1 (N=491 pairs)
Year 1 LR vs NR2 (N=608 pairs)
Year 2 DR vs NR1 (N=112 pairs)
Year 2 LR vs NR2 (N=155 pairs)

PPPM, per patient per month; LR, locoregional recurrence; DR, distant recurrence; NR, no recurrence; ER, emergency room. LR and DR cohorts were each matched 1:1 to their respective NR cohorts.

All-cause HCRU (Figure 2, Table 1)

- In year 1 post-recurrence, DR patients (491 pairs) had significantly higher incremental mean (SD) all-cause HCRU PPPM vs NR_1 (all P<0.001) by:
 - ✓ + 1.92 (4.24) ambulatory visits (outpatient visits + office visits)
 - ✓ + 0.23 (0.52) emergency room (ER) visits
 - ✓ + 0.12 (0.26) hospitalizations
 - ✓ + 1.23 (4.12) pharmacy fills
- Though more modest, LR patients (608 pairs) also had significantly higher HCRU PPPM vs NR_1 (all P<0.001) by:
 - ✓ + 1.45 (3.96) ambulatory visits
 - ✓ + 0.05 (0.29) ER visits
 - ✓ + 0.02 (0.13) hospitalizations
 - ✓ + 0.67 (3.65) pharmacy fills
- Year 2 showed a similar trend with significantly higher ambulatory visits and pharmacy fills for LR or DR vs respective NR cohorts (all P<0.05)

Figure 3. Incremental melanoma related HCRU PPPM for LR and DR vs respective NR in year 1 and 2 after recurrence

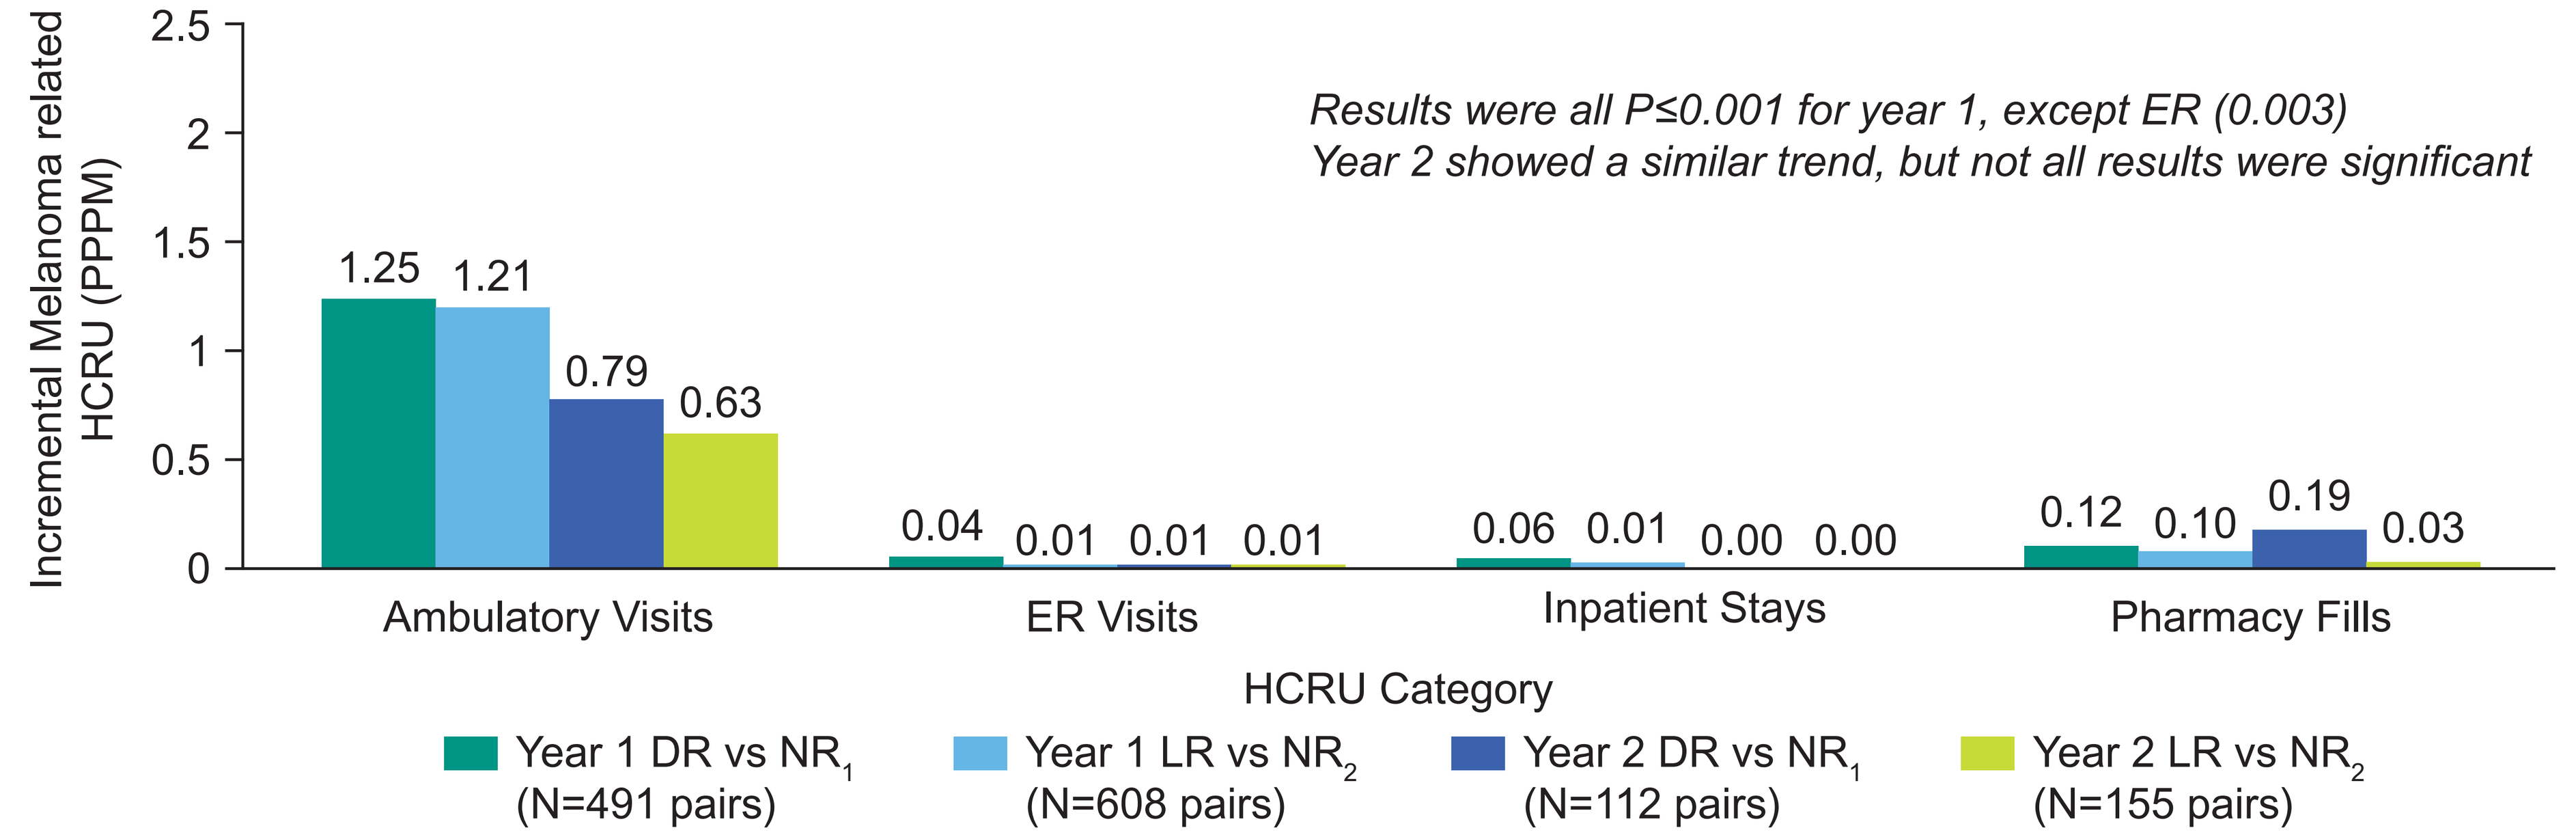
Incremental Melanoma related HCRU (PPPM)
2.5
2
1.5
1
0.5
0
1.25
1.21
0.79
0.63
0.04
0.01
0.01
0.01
0.06
0.01
0.00
0.00
0.12
0.10
0.19
0.03
Results were all P≤0.001 for year 1, except ER (0.003)
Year 2 showed a similar trend, but not all results were significant
Ambulatory Visits
ER Visits
Inpatient Stays
Pharmacy Fills
HCRU Category
Year 1 DR vs NR1 (N=491 pairs)
Year 1 LR vs NR2 (N=608 pairs)
Year 2 DR vs NR1 (N=112 pairs)
Year 2 LR vs NR2 (N=155 pairs)

PPPM, per patient per month; LR, locoregional recurrence; DR, distant recurrence; NR, no recurrence; ER, emergency room. LR and DR cohorts were each matched 1:1 to their respective NR cohorts.

Melanoma-related HCRU (Figure 3, Table 1)

- Melanoma-related incremental HCRU PPPM in year 1 was also significantly higher for DR vs NR_1 (all P<0.001) by:
 - ✓ + 1.25 (2.16) ambulatory visits
 - ✓ + 0.04 (0.12) ER visits
 - ✓ + 0.06 (0.14) hospitalizations
 - ✓ + 0.12 (0.48) pharmacy fills
- Melanoma-related HCRU PPPM for LR vs NR_2 in year 1 followed a similar trend and was significantly higher (all P<0.01) by:
 - ✓ + 1.21 (2.16) ambulatory visits
 - ✓ + 0.01 (0.11) ER visits
 - ✓ + 0.01 (0.06) hospitalizations
 - ✓ + 0.10 (0.54) pharmacy fills
- Year 2 showed a similar trend, but not all results were significant

Table 1. All-cause and melanoma-related HCRU in the first year after recurrence

All-cause HCRU in the first year after recurrence, PPPM, mean (SD)								
	DR (N = 491)	NR_1 (N = 491)	Difference (DR vs NR_1)	*P*-value	LR (N = 608)	NR_2 (N = 608)	Difference (LR vs NR_2)	*P*-value
Ambulatory visits	4.68 (3.43)	2.75 (2.95)	**1.92 (4.24)**	<0.001	4.30 (2.99)	2.85 (2.57)	**1.45 (3.96)**	<0.001
ER visits	0.30 (0.46)	0.08 (0.24)	**0.23 (0.52)**	<0.001	0.11 (0.24)	0.06 (0.17)	**0.05 (0.29)**	<0.001
Inpatient stays	0.15 (0.23)	0.02 (0.12)	**0.12 (0.26)**	<0.001	0.03 (0.11)	0.01 (0.06)	**0.02 (0.13)**	<0.001
Pharmacy fills	3.76 (3.22)	2.53 (2.68)	**1.23 (4.12)**	<0.001	2.96 (2.90)	2.29 (2.69)	**0.67 (3.65)**	<0.001
Length of inpatient stay[a]	2.66 (3.79)	2.01 (3.32)	-	-	1.57 (2.32)	1.87 (3.66)	-	-

Melanoma-related HCRU in the first year after recurrence, PPPM, mean (SD)								
	DR (N = 491)	NR_1 (N = 491)	Difference (DR vs NR_1)	*P*-value	LR (N = 608)	NR_2 (N = 608)	Difference (LR vs NR_2)	*P*-value
Ambulatory visits	2.26 (1.82)	1.01 (1.20)	**1.25 (2.16)**	<0.001	2.43 (1.81)	1.22 (1.25)	**1.21 (2.16)**	<0.001
ER visits	0.04 (0.11)	0.00 (0.02)	**0.04 (0.12)**	<0.001	0.02 (0.10)	0.01 (0.05)	**0.01 (0.11)**	0.003
Inpatient stays	0.06 (0.13)	0.00 (0.01)	**0.06 (0.14)**	<0.001	0.01 (0.05)	0.00 (0.02)	**0.01 (0.06)**	<0.001
Pharmacy fills	0.14 (0.44)	0.02 (0.18)	**0.12 (0.48)**	<0.001	0.16 (0.48)	0.06 (0.31)	**0.10 (0.54)**	<0.001
Length of inpatient stay[a]	2.29 (3.06)	3.45 (3.16)	-	-	1.92 (3.21)	1.65 (0.77)	-	-

[a]Among those with inpatient stay only. NR_1 and NR_2: No recurrence cohorts matched to DR and LR, respectively. PPPM, per patient per month; LR, locoregional recurrence; DR, distant recurrence; NR, no recurrence; ER, emergency room.

Limitations

Recurrence was identified using a claims-based algorithm and may have been misclassified or missed. As with all claims data, coding errors are possible, and findings may not generalize to uninsured populations. Missing stage information required a claims-based stage III proxy; however, sensitivity analyses excluding proxy-stage patients yielded results consistent with the primary analysis

Conclusions

Resected stage IIB–IIID melanoma patients experiencing recurrence had significantly higher HCRU than matched patients without recurrence, with the greatest incremental burden observed after distant recurrence. The incremental burden was most pronounced in the first year after recurrence and was evident across both all-cause and melanoma-related HCRU. These findings highlight the potential value of strategies that could reduce melanoma recurrence and the associated healthcare burden.

References

1. Rasmussen LA, et al., *Clin Epidemiol.* 2021;13:207-214. doi:10.2147/CLEP.S295844.
2. Jiang R, et al., *J Med Econ.* 2024;27(1):1279-1292. doi:10.1080/13696998.2024.2403283.
3. Jang S, et al., *Dermatol Ther (Heidelb).* 2020;10(5):985-999. doi:10.1007/s13555-020-00404-9.
4. Tarhini A, et al., *Cancer Manag Res.* 2019. doi:10.2217/cer-2019-0003.

Funding

This study was sponsored by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA, and Moderna, Inc., Cambridge, MA, USA, which are codeveloping intismeran autogene.

Copies of this poster obtained via the Quick Response (QR) Code or the Web link are for personal use only and may not be reproduced without written permission from the Congress or the author of this poster.

https://bit.ly/47ifJCx

Presented at The Academy of Managed Care Pharmacy (AMCP); October 26-29, 2026; San Diego, CA, USA.

Copyright © 2026 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.