Melanoma Recurrence Comes at a Cost: Real-World Economic Burden of Locoregional and Distant Recurrence in a Nationally Representative US Database

Ahmad A. Tarhini[1]; Salome S. Samant, MBBS, DHSc[2]; Yong Zhu, PhD[3]; Nicole Engel-Nitz, PhD[3]; Dmitri O. Grebennik, MD[2]; Shujing Zhang, PhD[2]; Laura Becker, MS[3]; Arunima Sachdev, MA[3]; Shiqiang Li, MS[3]; Sameer R. Ghate, PhD[2]

[1]H. Lee Moffitt Cancer Center and Research Institute, Tampa, Florida, USA;
[2]Merck & Co., Inc., Rahway, NJ, USA; [3]Optum, Eden Prairie, MN, USA

Background

- Patients with resected stage IIB/C or stage III melanoma remain at risk for locoregional recurrence (LR) and distant recurrence (DR), which can lead to substantial downstream healthcare utilization and costs.[1-4] Contemporary real-world estimates are needed to quantify the economic consequences of recurrence across payer types in the current treatment era

Objective

- To estimate incremental all-cause and melanoma-related healthcare costs associated with LR and DR versus no recurrence (NR) among adults with resected stage IIB–IIID cutaneous melanoma in the US

Methods

- Retrospective matched cohort study using nationally representative Optum Market Clarity claims linked to electronic health record data with costs standardized across all payer types (Commercial, Medicare, and Medicaid)
- Adults diagnosed from July 1, 2019 – May 31, 2025 with stage IIB–IIID cutaneous melanoma (or claims-based proxy for stage III) were included if they underwent melanoma-related resection within 120 days of diagnosis, with the last date of resection considered index date 1 (ID_1). Patients with other primary cancers/pregnancy/clinical trial participation were excluded (**Figure 1**)
- LR and DR were identified using prespecified claims-based algorithms developed based on literature[1-4]; LR patients who later progressed to DR were included in both analyses but censored 30 days before DR onset for the LR analysis
- The earliest recurrence date of LR or DR was set as index date 2 (ID_2) for LR or DR analysis respectively
- Patients with LR and DR were separately matched 1:1 to NR controls using propensity score matching based on baseline patient characteristics and healthcare costs. Matched NR controls (NR_1 and NR_2) were assigned separately to each DR and LR cohort and were assigned an index date (ID_2) so the time between ID_1 and ID_2 is the same in matched pairs
- All-cause and melanoma-related costs were reported per patient per month (PPPM) in 2024 US dollars during year 1 (30 days prior to 11 months after recurrence) and year 2 (11 months to 23 months after recurrence). Annualized cost savings were also estimated

Figure 1. Patient attrition

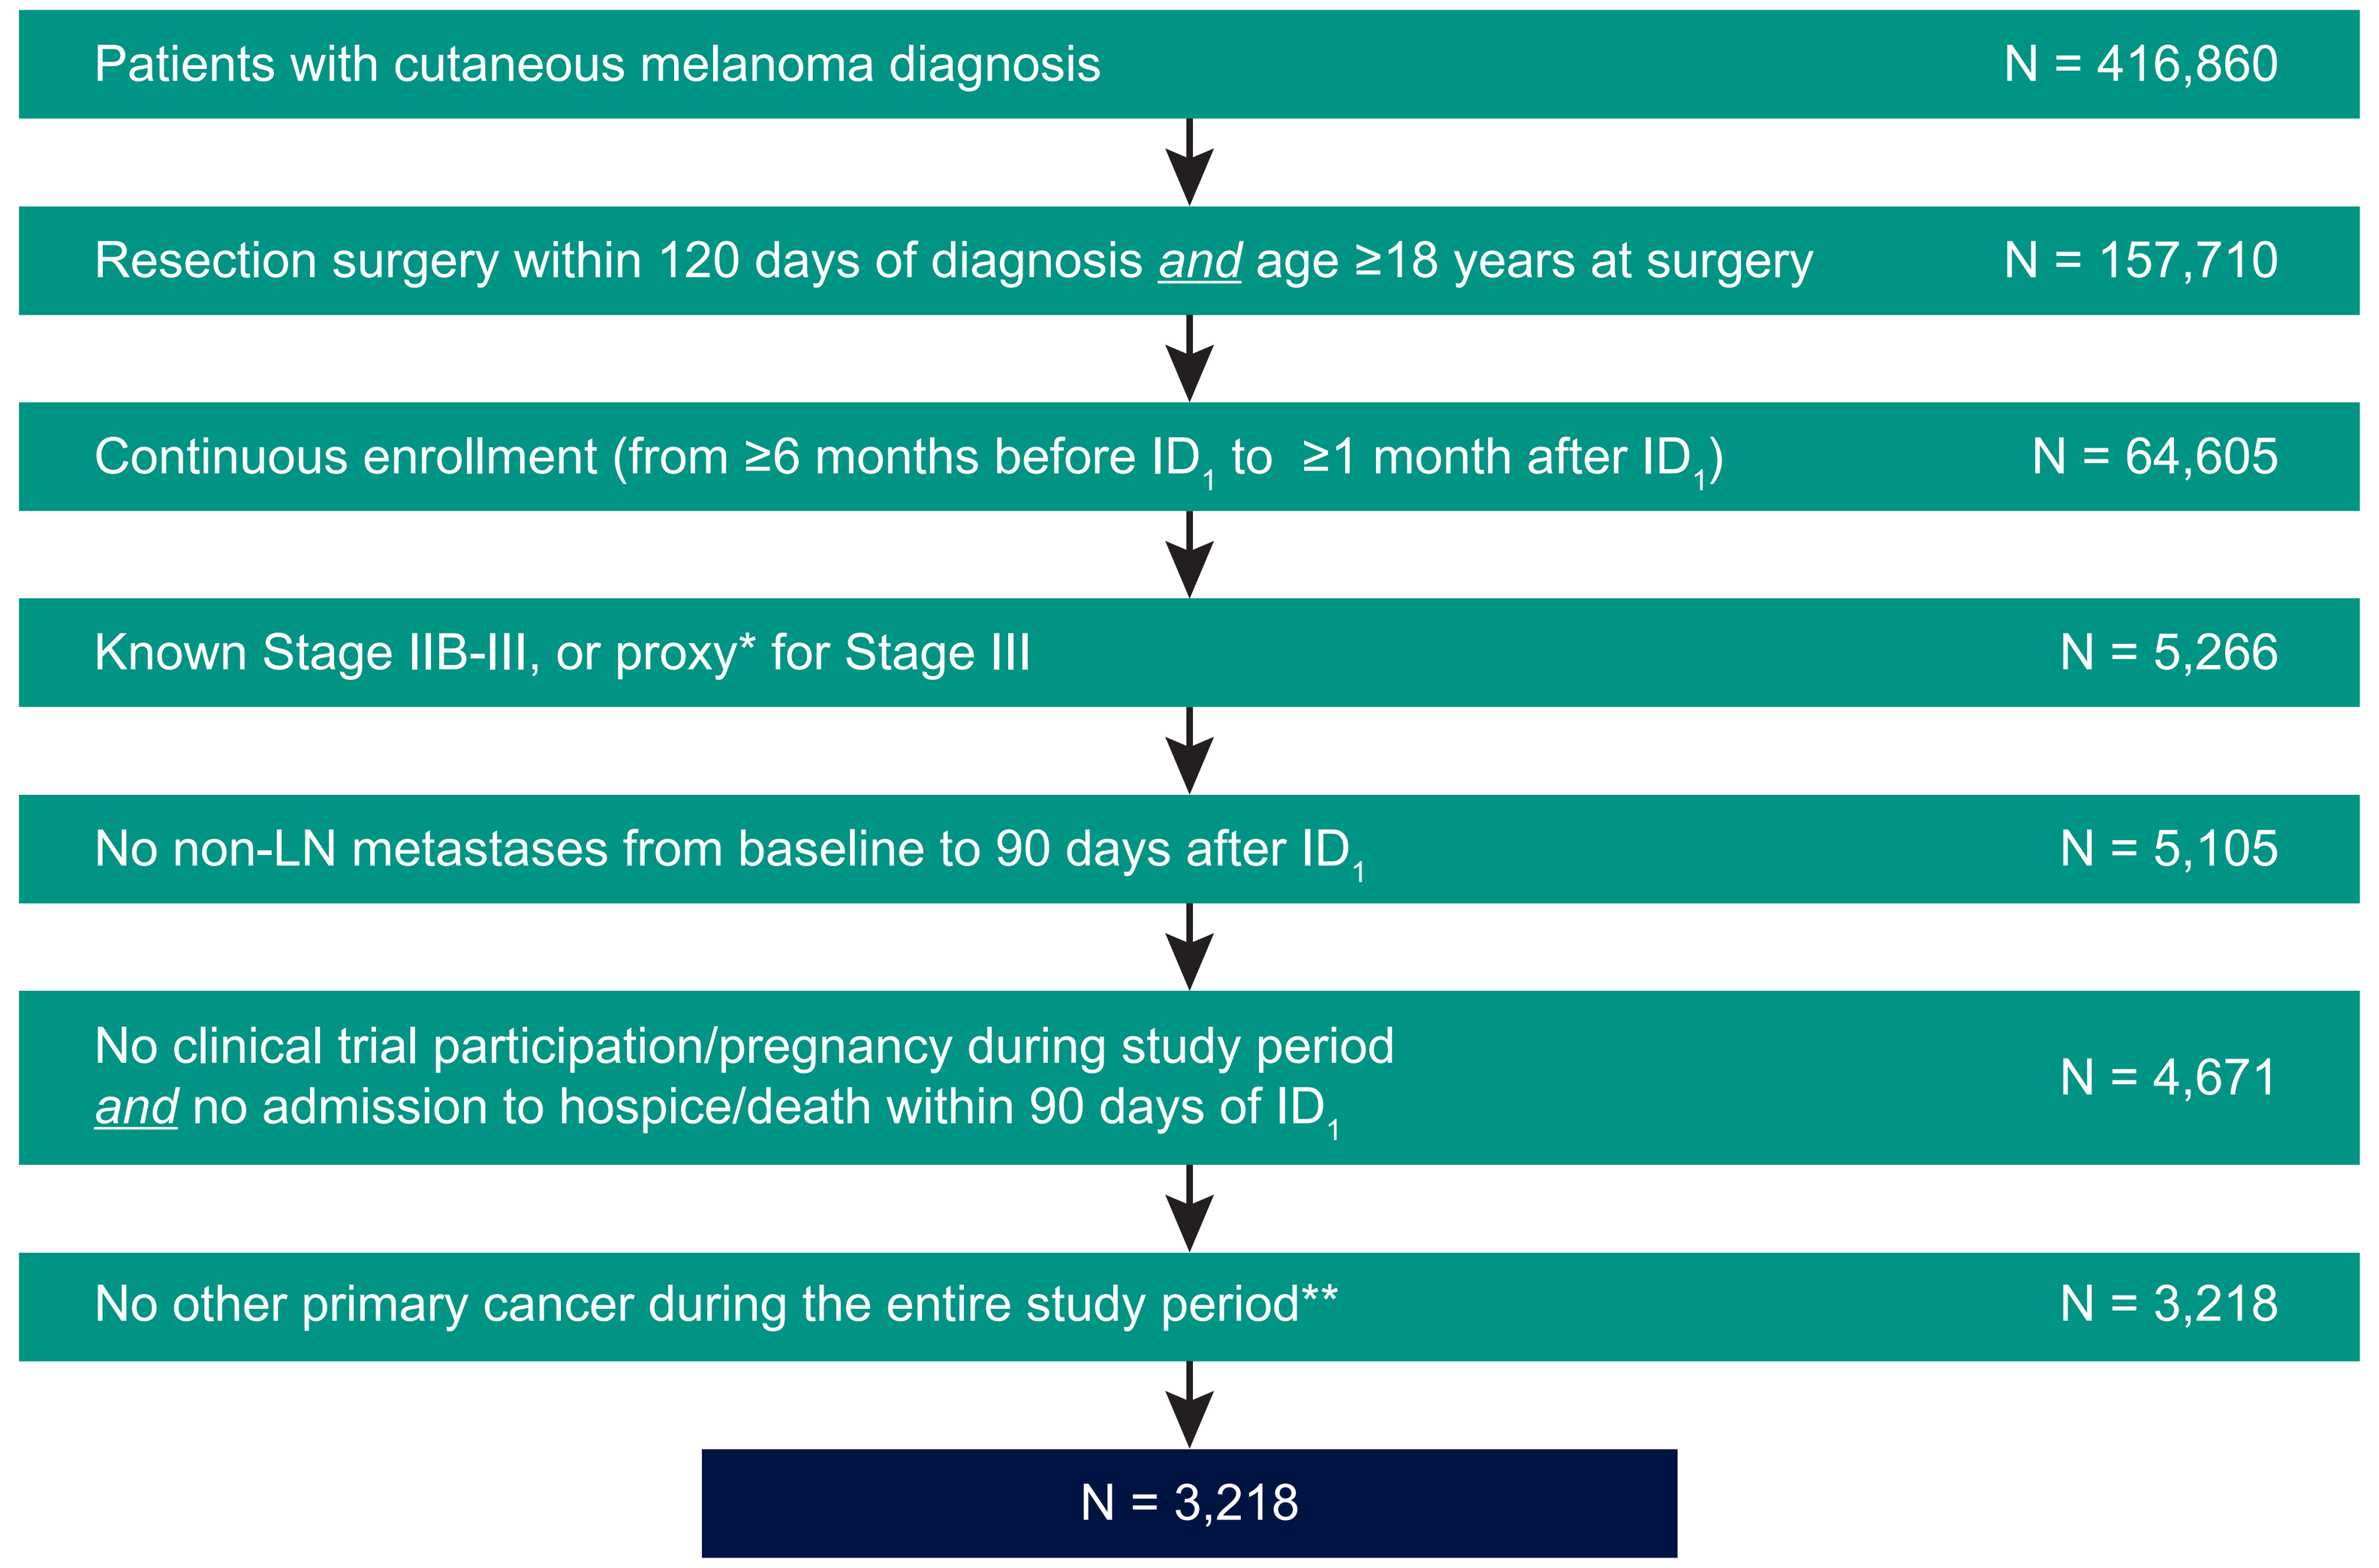
Patients with cutaneous melanoma diagnosis N = 416,860
Resection surgery within 120 days of diagnosis *and* age ≥18 years at surgery N = 157,710
Continuous enrollment (from ≥6 months before ID_1 to ≥1 month after ID_1) N = 64,605
Known Stage IIB-III, or proxy* for Stage III N = 5,266
No non-LN metastases from baseline to 90 days after ID_1 N = 5,105
No clinical trial participation/pregnancy during study period *and* no admission to hospice/death within 90 days of ID_1 N = 4,671
No other primary cancer during the entire study period** N = 3,218
N = 3,218

*For Stage III Proxy: Have a lymph node (LN) excision surgery within 120 days following diagnosis and had no non-lymph node metastasis in the 2-months prior to diagnosis through 2-months following ID1 (resection) AND had LN metastasis in that period; **Except squamous cell or basal cell carcinomas or carcinomas in situ, for the entire study period from the beginning of baseline period to end of follow-up period or death.

Results

Study population:

- After applying inclusion and exclusion criteria, 3,218 patients were identified; 91% were identified as stage III using proxy, with known stage IIB/C and known III accounting for remaining 2.8% and 6.1%, respectively
- Overall, 1,000 patients had recurrence, including 616 with LR and 521 with DR. After matching, year 1 analyses included 491 DR–NR_1 pairs and 608 LR–NR_2 pairs; year 2 analyses included 112 DR–NR_1 pairs and 155 LR–NR_2 pairs

Figure 2. All-cause mean total healthcare costs for matched pairs in year 1 or year 2 after recurrence

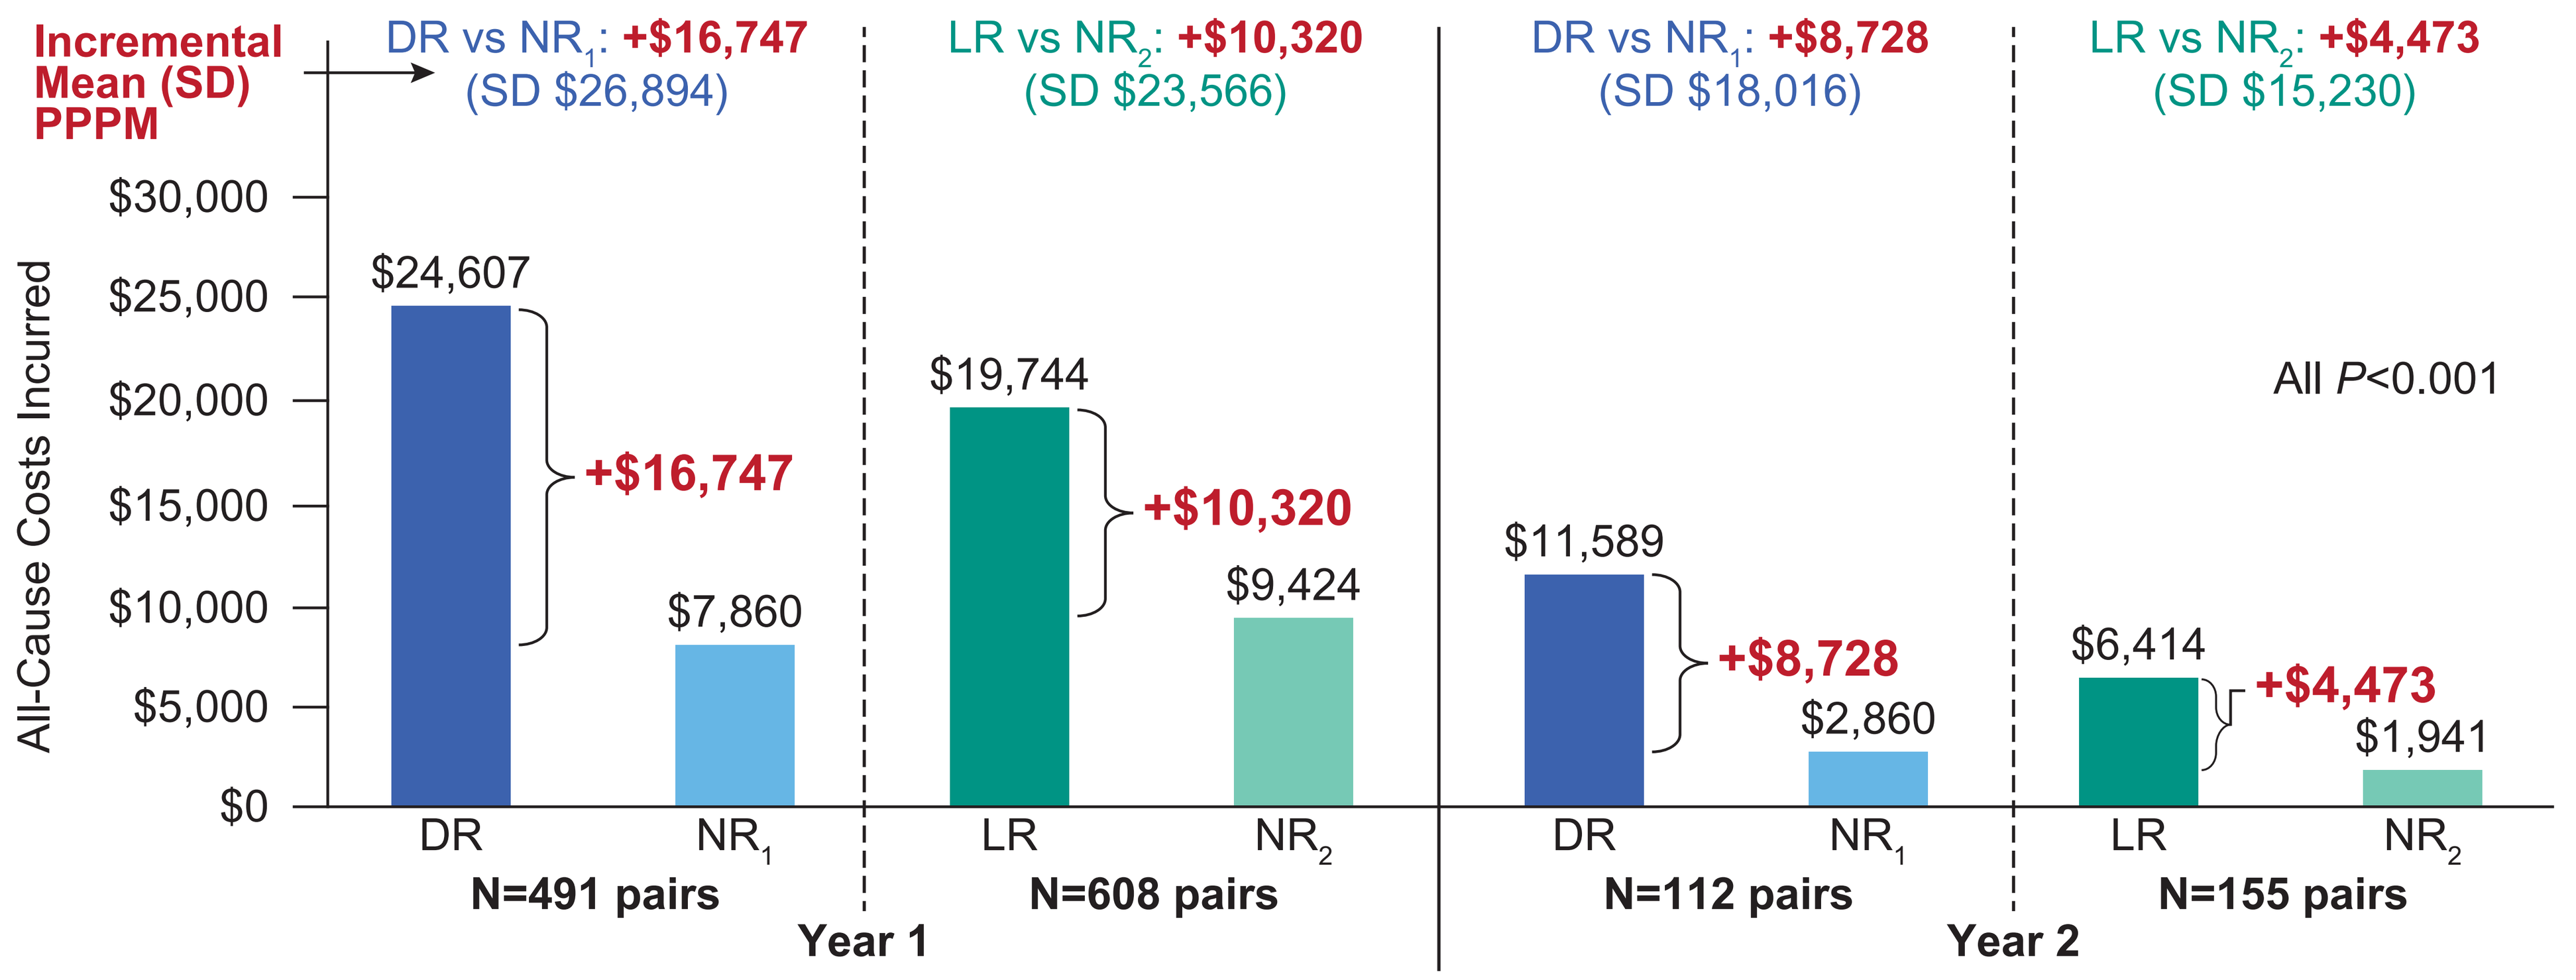
Incremental Mean (SD) PPPM
DR vs NR_1: +$16,747 (SD $26,894)
LR vs NR_2: +$10,320 (SD $23,566)
DR vs NR_1: +$8,728 (SD $18,016)
LR vs NR_2: +$4,473 (SD $15,230)
All-Cause Costs Incurred
$30,000
$25,000
$20,000
$15,000
$10,000
$5,000
$0
$24,607
+$16,747
$7,860
$19,744
+$10,320
$9,424
$11,589
+$8,728
$2,860
$6,414
+$4,473
$1,941
All P<0.001
DR
NR_1
LR
NR_2
DR
NR_1
LR
NR_2
N=491 pairs
N=608 pairs
N=112 pairs
N=155 pairs
Year 1
Year 2

PPPM: Per patient per month; DR: Distant recurrence; LR: Locoregional recurrence; NR_1 and NR_2: No recurrence cohorts matched to DR and LR, respectively.

Figure 3. Incremental all-cause costs in year 1 after recurrence, by site of service, PPPM, mean (SD)

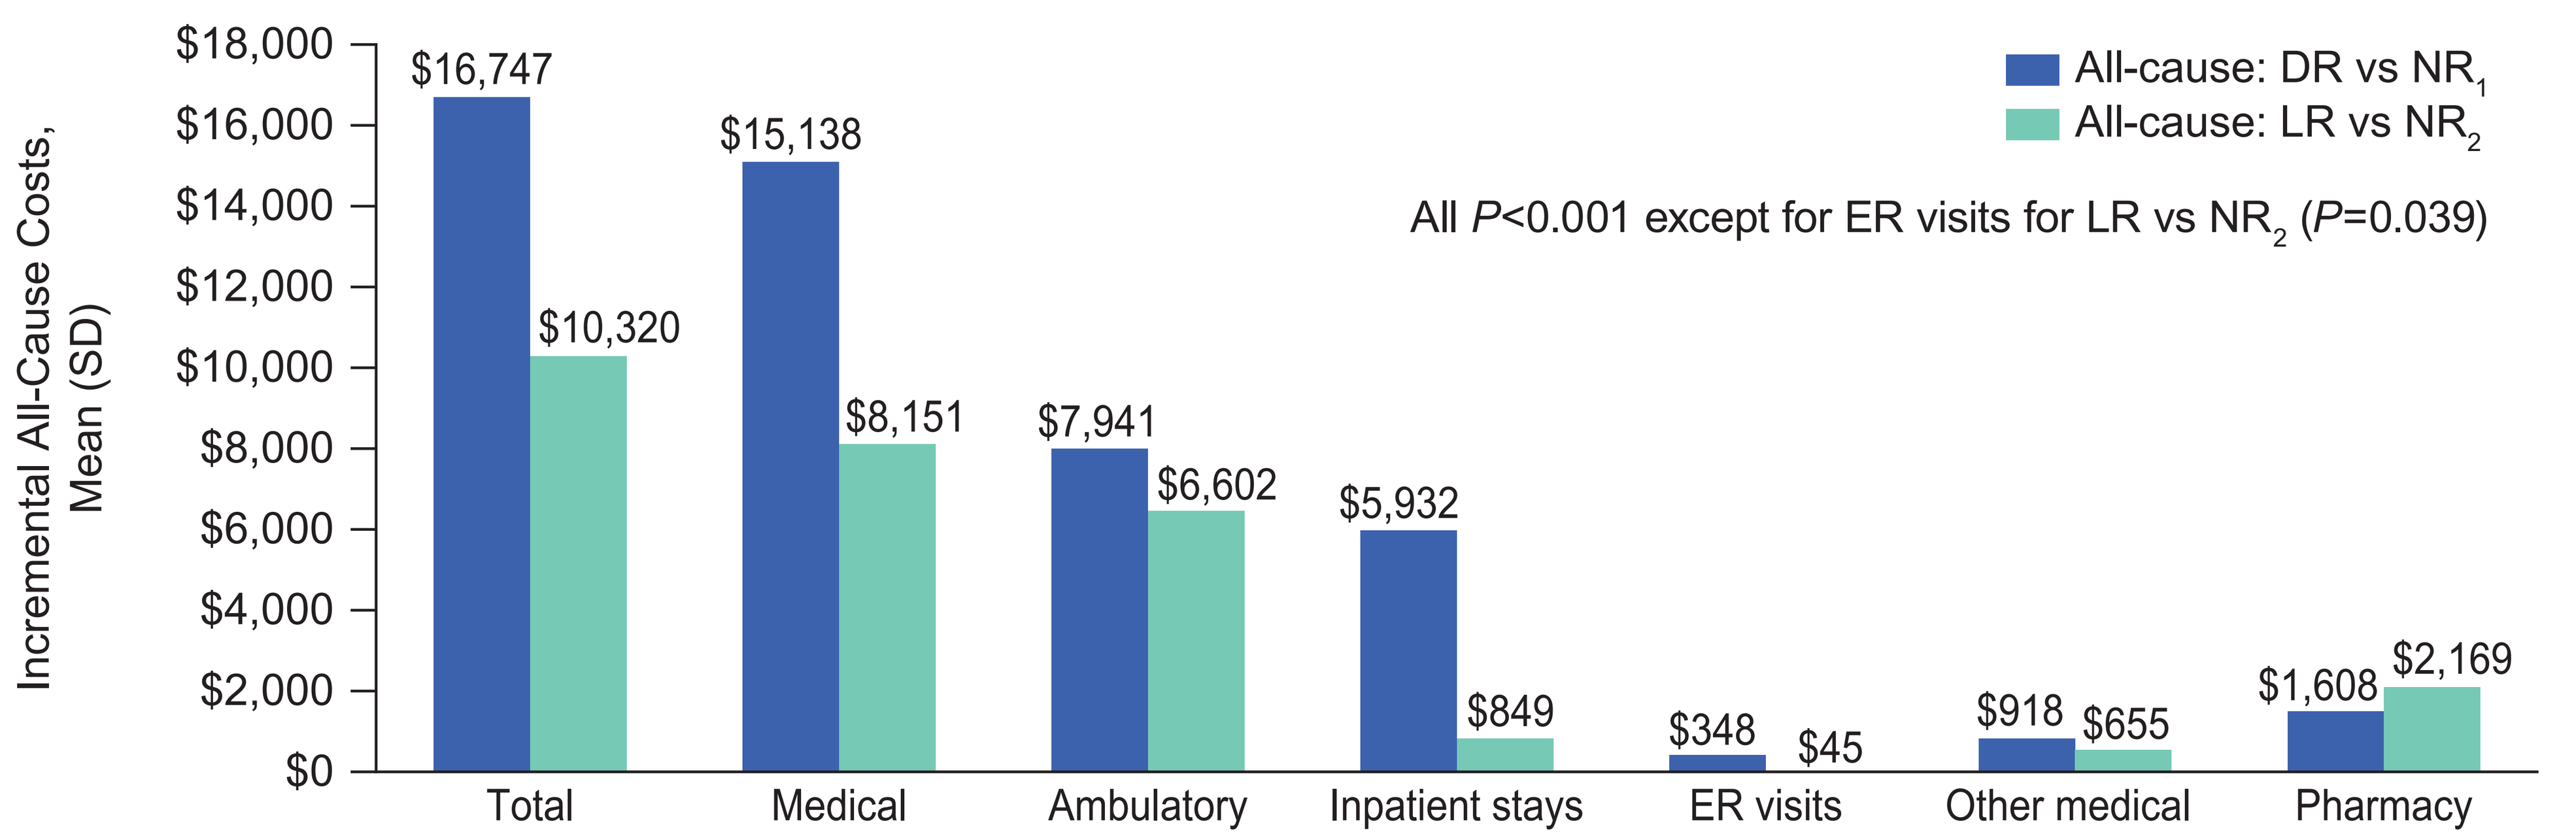
Incremental All-Cause Costs, Mean (SD)
$18,000
$16,000
$14,000
$12,000
$10,000
$8,000
$6,000
$4,000
$2,000
$0
All-cause: DR vs NR_1
All-cause: LR vs NR_2
All P<0.001 except for ER visits for LR vs NR_2 (P=0.039)
$16,747
$10,320
$15,138
$8,151
$7,941
$6,602
$5,932
$849
$348
$45
$918
$655
$1,608
$2,169

	Total	Medical	Ambulatory	Inpatient stays	ER visits	Other medical	Pharmacy
DR vs NR_1	$16,747 (26,894)	$15,138 (25,958)	$7,941 (17,091)	$5,932 (19,253)	$348 (1,261)	$918 (3,635)	$1,608 (8,637)
LR vs NR_2	$10,320 (23,566)	$8,151 (20,552)	$6,602 (18,821)	$849 (5,876)	$45 (539)	$655 (4,368)	$2,169 (13,484)

Total costs = Medical costs (Ambulatory + ER visits + Inpatient + Other medical) + Pharmacy costs (prescription-only claims). Immunotherapy was mainly captured under medical costs depending on the setting; ER: Emergency room; PPPM: Per patient per month; DR: Distant recurrence; LR: Locoregional recurrence; NR_1 and NR_2: No recurrence cohorts matched to DR and LR, respectively; SD: Standard deviation.

All-cause costs:

- In year 1 post-recurrence, mean all-cause total healthcare costs were substantially higher for DR versus NR_1 patients (+$16,747 PPPM; $24,607 vs $7,860; P<0.001) and for LR versus NR_2 patients (+$10,320 PPPM; $19,744 vs $9,424; P<0.001) (Figure 2)
- In year 2 post-recurrence, differences persisted but attenuated, with incremental costs of +$8,728 PPPM for DR and +$4,473 PPPM for LR versus respective NR cohorts (both P<0.001)

Ambulatory care (outpatient and office visits) was a major contributor to incremental costs for both DR and LR. However, inpatient costs contributed disproportionately to the higher overall economic burden (**Figure 3**)

Melanoma-related costs:

- Year 1 melanoma-related costs were also significantly higher for DR vs NR_1 patients (+$10,953 PPPM; $16,148 vs $5,195; P<0.001) and for LR vs NR_2 patients (+$8,493 PPPM; $15,423 vs $6,930; P<0.001)
- In year 2, melanoma-related incremental costs remained elevated at +$6,784 PPPM for DR vs NR_1 and +$4,290 PPPM for LR vs NR_2 (both P<0.001)

Potential annual cost savings:

- When year 1 post-recurrence incremental PPPM all-cause costs were annualized across 100 matched pairs each by multiplying the PPPM incremental costs by 12, recurrence was associated with an estimated excess cost of approximately $20.1 million for DR and $12.4 million for LR compared with respective matched NR controls. These illustrative estimates highlight the potential economic value of interventions that reduce recurrence risk, particularly distant recurrence

Figure 4. Potential annualized recurrence-associated all-cause excess costs across 100 patient pairs

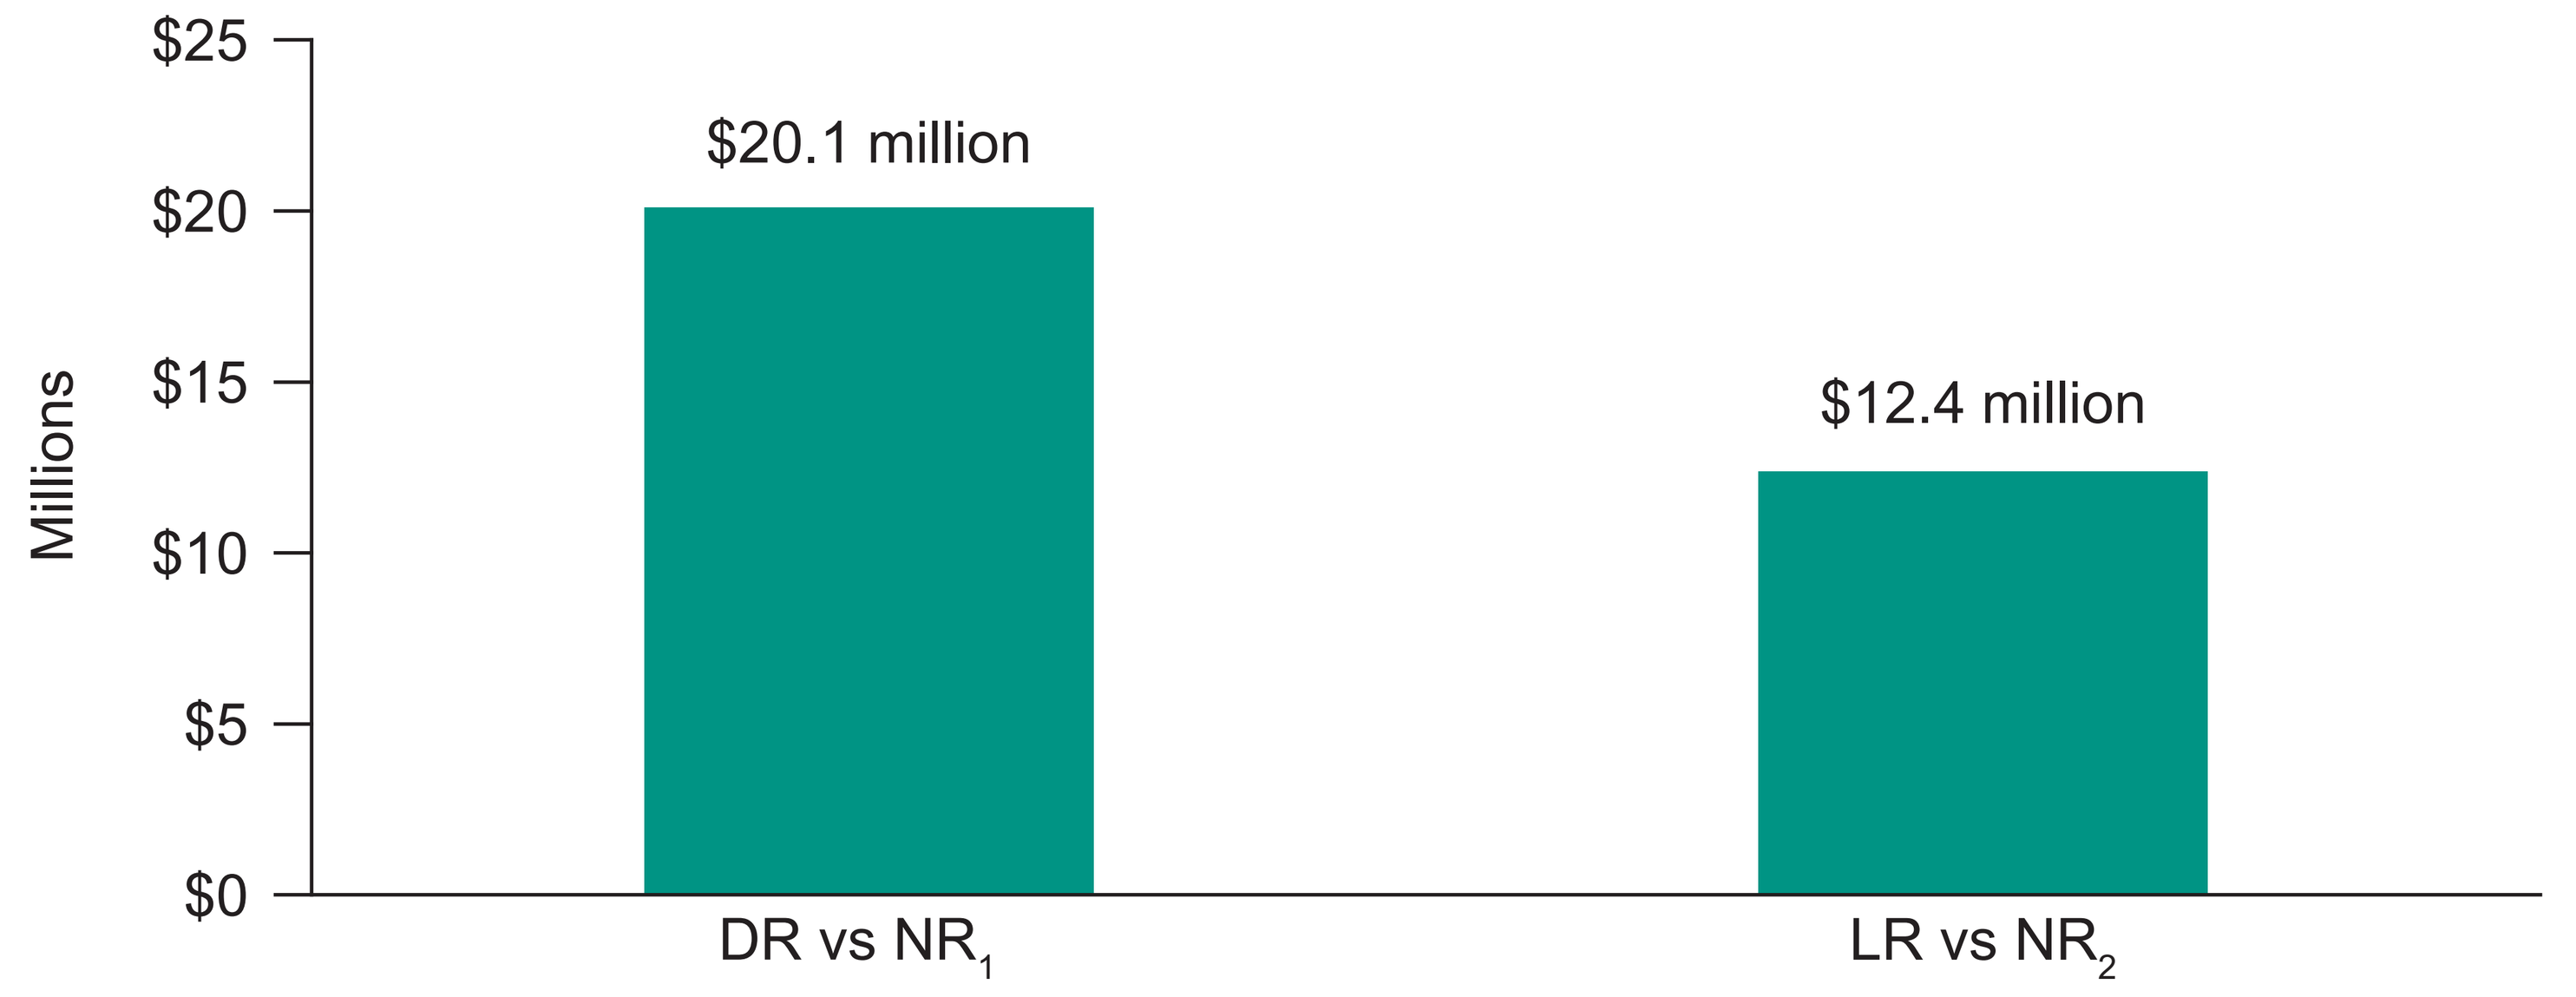
Millions
$25
$20
$15
$10
$5
$0
$20.1 million
$12.4 million
DR vs NR_1
LR vs NR_2

Incremental PPPM costs across 100 matched patient pairs each were annualized to estimate the excess cost versus no recurrence (ie, the potential savings if recurrence-associated excess costs were avoided). DR: Distant recurrence; LR: Locoregional recurrence; NR: No recurrence.

Limitations

- Recurrence was identified using a claims-based algorithm, leading to some recurrence events possibly being misclassified or missed. As with all claims data, misclassification and coding inaccuracies may occur. Results may also not be generalizable to uninsured patients. Missing stage data required a claims-based stage III proxy; however sensitivity analyses excluding proxy-stage patients were consistent with the main analysis

Conclusions

- In resected stage IIB-IIID melanoma, both LR and DR are associated with marked increases in healthcare costs compared with NR, with DR reflecting the greatest economic burden
- Interventions that effectively reduce recurrence risk may offer not only clinical benefit but also meaningful economic value to payers

References

1. Rasmussen LA, et al., *Clin Epidemiol.* 2021;13:207-214. doi:10.2147/CLEP.S295844.
2. Jiang R, et al., *J Med Econ.* 2024;27(1):1279-1292. doi:10.1080/13696998.2024.2403283.
3. Jang S, et al., *Dermatol Ther (Heidelb).* 2020;10(5):985-999. doi:10.1007/s13555-020-00404-9.
4. Tarhini A, et al., *Cancer Manag Res.* 2019. doi:10.2217/cer-2019-0003.

Funding

This study was sponsored by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA, and Moderna, Inc., Cambridge, MA, USA, which are codeveloping intismeran autogene.

Copies of this poster obtained via the Quick Response (QR) Code or the Web link are for personal use only and may not be reproduced without written permission from the Congress or the author of this poster.

https://bit.ly/4yVke1H

Presented at The Academy of Managed Care Pharmacy (AMCP); October 26-29, 2026; San Diego, CA, USA.

Copyright © 2026 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.